A Phase 1/2 First-in-Human Trial of Oral SRA737 (a Chk1 Inhibitor) in Subjects with Advanced Cancer

Abstract #3094

Authors and Affiliations

Ruth Plummer[1], Rebecca Kristeleit[2], Elena Cojocaru[3], Noor Haris[1], Louise Carter[4], Robert Jones[5], Sarah Blagden[6], Jeff Evans[7], Tobias Arkenau[8], Debashis Sarker[9], Sarah Danson[10], Stefan Symeonides[11], Harriet Walter[12], Joanita Murangira Ocen[5], Manreet Randhawa[7], Christian Hassig[13], Mark Kowalski[13], Bryan Strouse[13], Ines Verdon[13], Andrew Dye[13], Michael Woolliscroft[13], Udai Banerji[3]

[1]Freeman Hospital, Newcastle, UK; [2]University College Hospital, London, UK; [3]Royal Marsden Hospital, London, UK; [4]The Christie Hospital, Manchester, UK; [5]Velindre Cancer Centre, Cardiff, UK; [6]Oxford University Hospital, Oxford, UK; [7]The Beatson West of Scotland Cancer Center, Glasgow, UK; [8]Sarah Cannon Research Institute, London, UK; [9]Guy's Hospital, London, UK; [10]Weston Park Hospital, Sheffield, UK; [11]Western General Hospital, Edinburgh, UK; [12]Leicester Royal Infirmary, Leicester, UK; [13]Sierra Oncology, Vancouver, Canada

Introduction

SRA737 is a potent, highly selective and orally-bioavailable small molecule inhibitor of Checkpoint kinase 1 (Chk1).

Chk1 is a serine/threonine protein kinase in the **DNA Damage Response (DDR)** network that is critically important in reducing elevated replication stress in certain tumor cells.

Replication stress (RS) is manifested by the slowing and stalling of replication forks which results in DNA that is prone to damage. Increased RS results in genomic instability, which affords survival advantages to tumor cells, however, if not properly managed, can result in extensive DNA damage and cell death.

Consequently, tumor cells increase **reliance on Chk1 to manage elevated intrinsic RS**. It is hypothesized that cancer cells with higher RS may have increased sensitivity to Chk1 inhibitor therapy.

Intrinsic sources of RS can include genetic alterations in **tumor suppressors, oncogenes or DNA damage repair genes**.

Tumors harboring defects in these functional gene networks are hypothesized to have higher levels of intrinsic RS due to dysregulated cell cycle control, increased proliferation demands and increased genomic instability **(Figure 1)**.

This signal-seeking Phase 1/2 study (NCT02797964) was designed to investigate the safety and tolerability of continuous, daily dosing of SRA737, as well as to evaluate preliminary anti-tumor activity in tumors with genetic alterations predicted to confer increased intrinsic RS and Chk1i sensitivity.

Prospective genetic screening was performed to identify and select subjects harboring two or more of these genetic alterations. The study was designed as a broad survey in order to explore the association between various sources of intrinsic RS and SRA737 monotherapy anti-tumor activity and to delineate potential genetic signatures and/or tumor indications that might warrant additional therapeutic investigation.

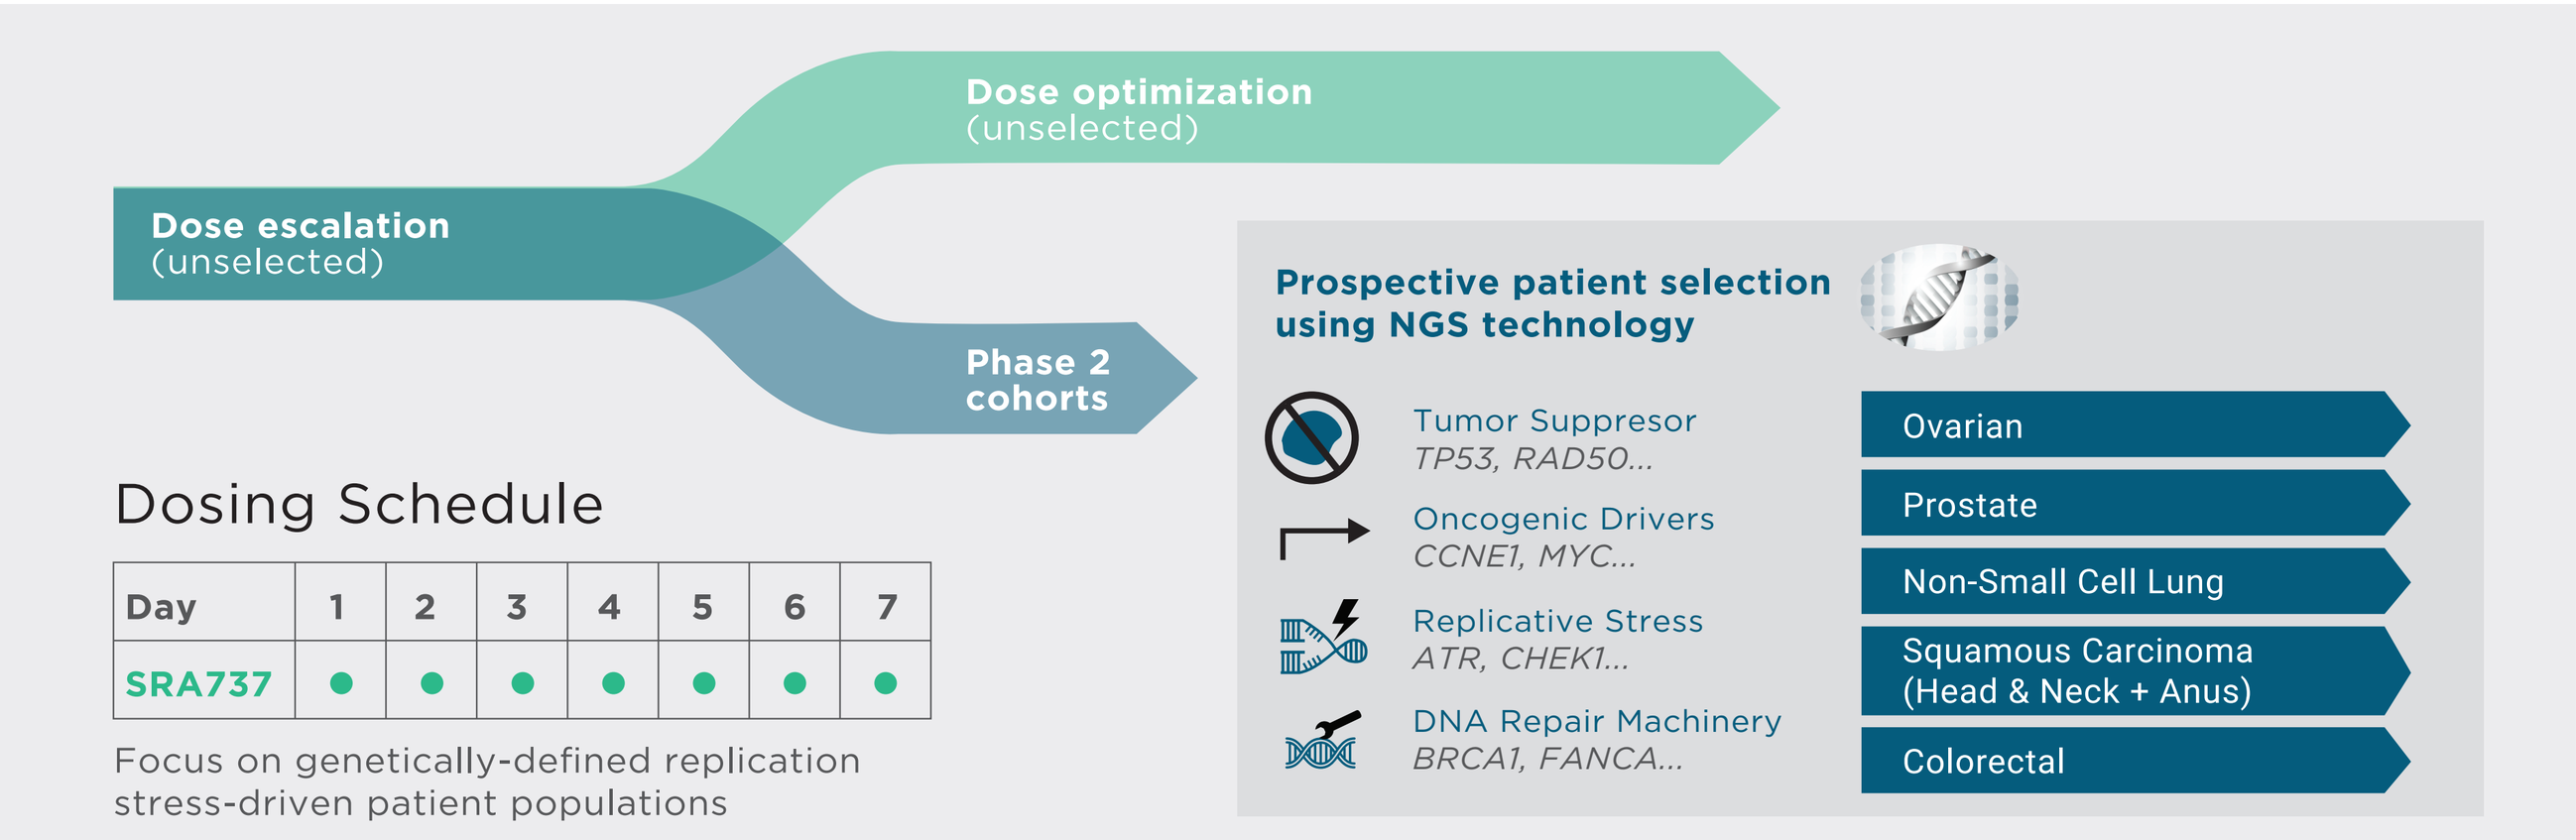

Dose optimization (unselected)
Dose escalation (unselected)
Phase 2 cohorts
Dosing Schedule
Day 1 2 3 4 5 6 7
SRA737
Focus on genetically-defined replication stress-driven patient populations
Prospective patient selection using NGS technology
Tumor Suppressor TP53, RAD50...
Oncogenic Drivers CCNE1, MYC...
Replicative Stress ATR, CHEK1...
DNA Repair Machinery BRCA1, FANCA...
Ovarian
Prostate
Non-Small Cell Lung
Squamous Carcinoma (Head & Neck + Anus)
Colorectal

Strategy & Rationale

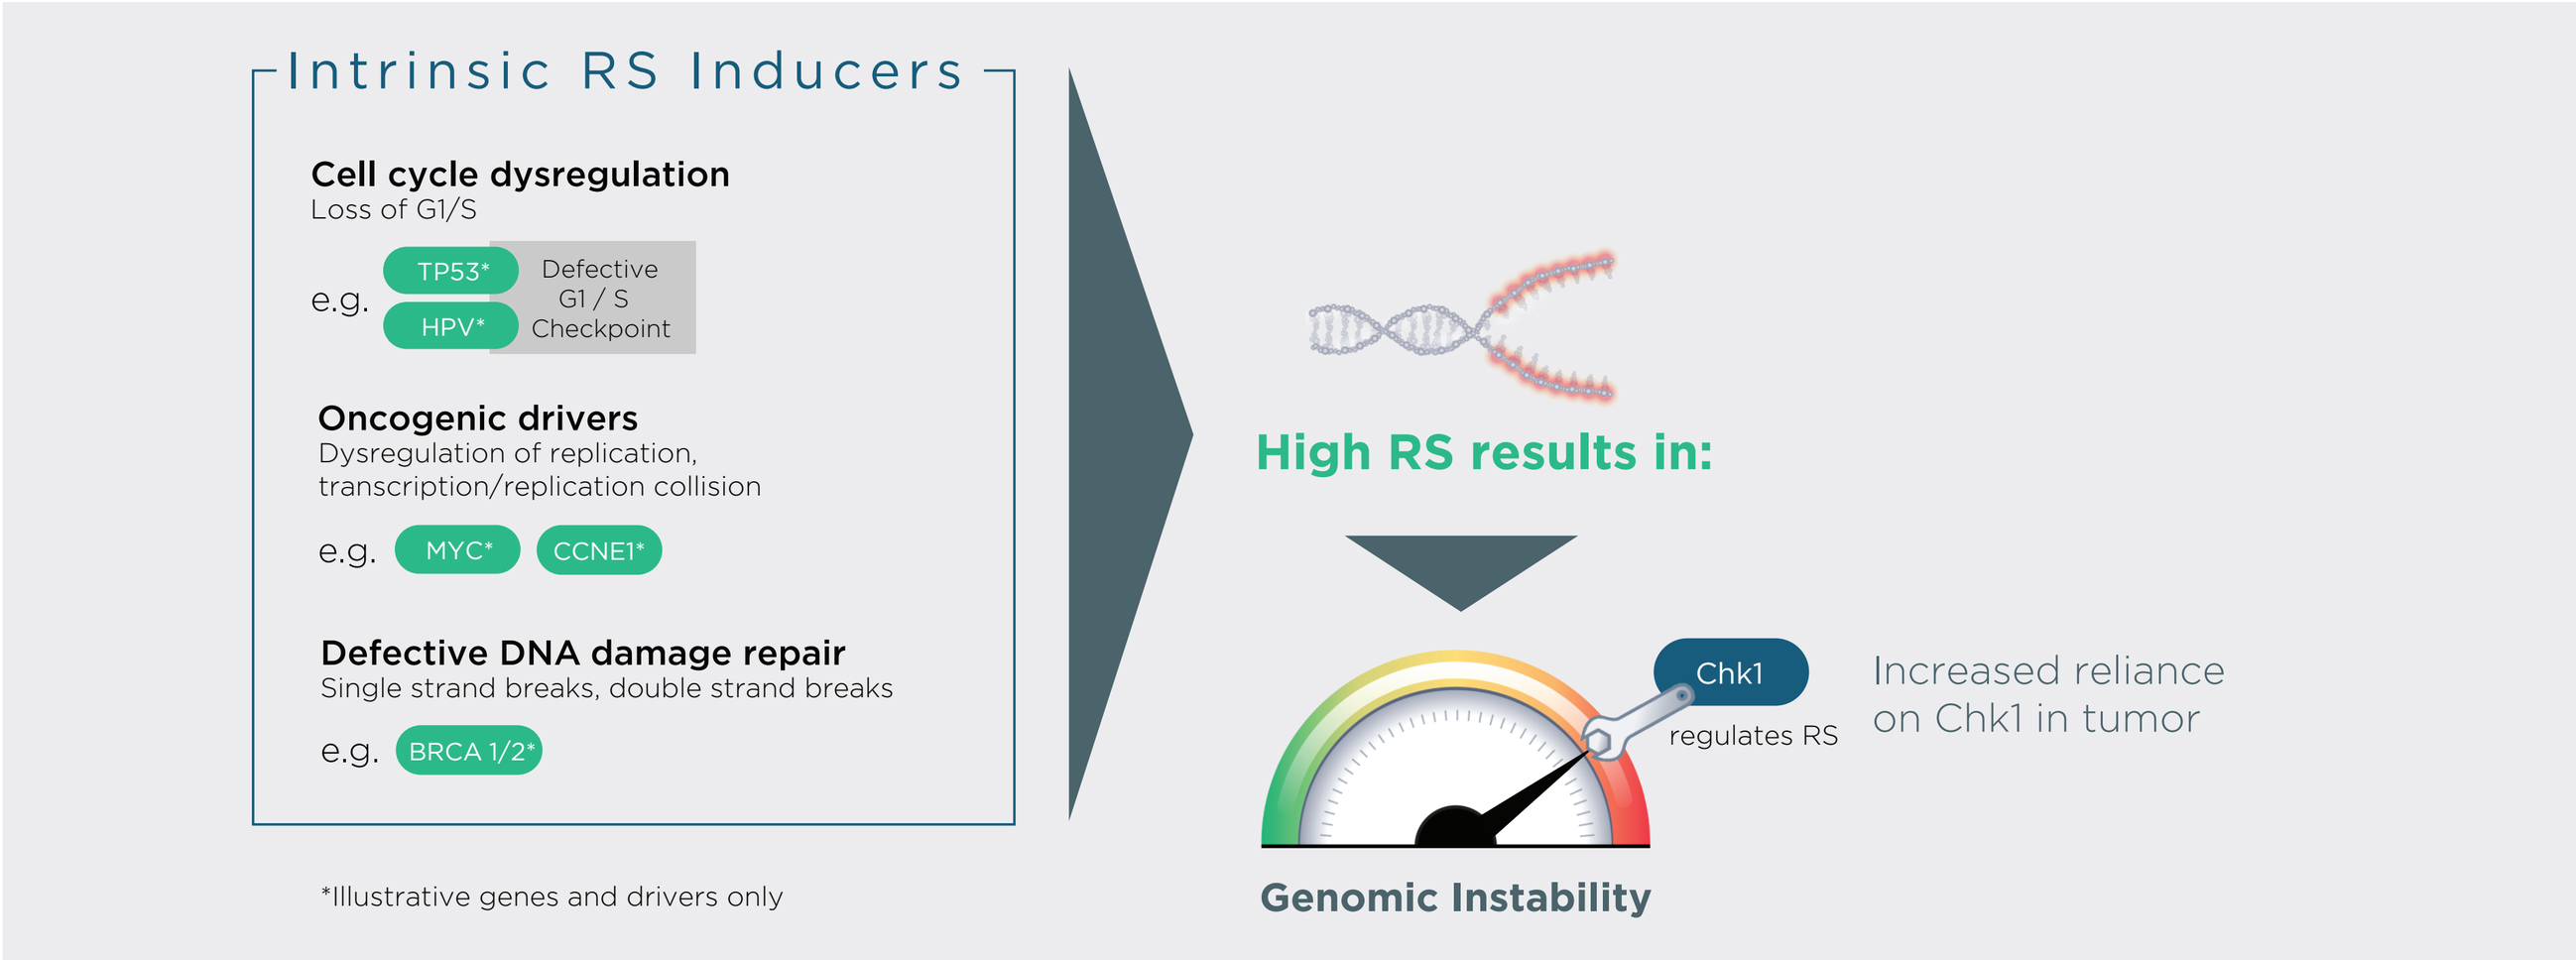

Intrinsic RS Inducers
Cell cycle dysregulation
Loss of G1/S
e.g. TP53* HPV* Defective G1 / S Checkpoint
Oncogenic drivers
Dysregulation of replication, transcription/replication collision
e.g. MYC* CCNE1*
Defective DNA damage repair
Single strand breaks, double strand breaks
e.g. BRCA 1/2*
*Illustrative genes and drivers only
High RS results in:
Chk1
regulates RS
Increased reliance on Chk1 in tumor
Genomic Instability

Figure 1. Intrinsic Inducers of RS Increase Tumor Cell Reliance on Chk1.

RS-driver genes can be divided into several functional categories including G1/S tumor suppressors, oncogenes and DNA repair genes. Research in the DDR field has implicated mutations in G1/S guardian genes, including *RB1*, *TP53* and genes functioning in these pathways, as potentially contributing to intrinsic RS. Certain viral infections that impact these same pathways, e.g. HPV, have also been implicated in increasing cellular RS. In addition, activating mutations in several oncogenes, including *MYC*, *CCNE1* and others, have been suggested to dysregulate replication origin firing and transcriptional programs resulting in elevated RS. Similarly, tumor genetic alterations in certain DNA repair factors, such as those in the Fanconi Anemia and BRCA pathways, have been demonstrated to compromise replication fork stability or repair of damaged forks, exacerbating intrinsic RS.

Methods

The Dose Escalation phase employed an accelerated titration design starting at 20 mg SRA737, administered QD orally in 28-day cycles. Incremental dose escalations in single-subject cohorts were followed by a rolling-6 design once SRA737-related ≥ Grade 2 toxicity was observed during Cycle 1.

The Cohort Expansion phase was contemporaneously initiated when circulating plasma concentrations of SRA737 exceeded the minimum effective concentration of SRA737 modelled from murine efficacy studies. Thereafter, experience gained in the ongoing Dose Escalation phase informed dose selection for expansion cohorts.

The Cohort Expansion phase enrolled subjects with genetically defined tumors that harbored genomic alterations hypothesized to confer sensitivity to Chk1 inhibition, which were prospectively selected by next-generation sequencing (FoundationOne). Subjects with the following tumors were eligible for enrollment: ia) high grade serous ovarian cancer (HGSOC), ib) HGSOC putatively enriched for *CCNE1* gene network amplification, ii) colorectal cancer (CRC), iii) metastatic castration-resistant prostate cancer (mCRPC), iv) non-small cell lung cancer (NSCLC), and v) squamous cell carcinomas (head and neck (HNSCC); anus (SCCA)).

This signal seeking Phase 1/2 study was generally conducted in specialty Phase 1 cancer units. Subjects ≥ 18 years of age with an ECOG performance status of 0–1, measurable disease (per RECIST v1.1) and archival tumor tissue (or willingness to consent to a biopsy) were eligible to participate in the study. For expansion cohorts, subjects must have received at least one prior regimen for advanced/metastatic disease.

Subject Characteristics and Dose Evaluation

In the Dose Escalation phase, 18 subjects received SRA737 across 9 dose level cohorts, ranging from 20 to 1300 mg QD. Of these subjects, 3 experienced DLTs (inability to receive 75% of the planned dose); 2 at 1300 mg QD due to gastrointestinal intolerability and 1 at 500 mg BID due to thrombocytopenia. The maximum tolerated dose (MTD) was established at 1000 mg QD or 500 mg BID.

Enrollment of the Cohort Expansion phase was initiated at 600 mg SRA737. Based on overall tolerability, including particularly common gastrointestinal events (nausea, vomiting, and diarrhea), the recommended dose to be employed in the expansion cohorts was determined to be 800 mg QD (RP2D).

In the Cohort Expansion phase, of the 512 subjects prospectively identified, 355 were screened for genetic alterations associated with Chk1 sensitivity. Of these subjects 237 (67%) met genetic eligibility criteria, and 94 were treated in expansion cohorts across six tumor types.

Characteristic	Overall (Escalation + Expansion)	Tumor types of Interest (Expansion)*: HGSOC (inc. CCNE1 enriched)**	CRC**	NSCLC	mCRPC**
Number of subjects treated	107	38	27	10	15
Age / years (min, max)	64 (38, 86)	60 (39, 86)	64 (38, 79)	66 (50, 75)	68 (54, 80)
Gender (M/F)	46 (43%) / 61 (57%)	0 / 38 (100%)	17 (63%) / 10 (37%)	4 (40%) / 6 (60%)	15 (100%) / 0
WHO performance status (PS0 / PS1 / PS unknown)	39 / 66 / 2	14 / 23 / 1	16 / 10 / 1	3 / 7 / -	4 / 11 / -
Prior systemic therapy regimens; mean (min, max)***	4.2 (1, 10)	4.7 (2, 10)	3.5 (2, 5)	3.2 (2, 6)	5.7 (2, 10)
Treatment delay from consent to C1D1; median (min, max)	61 (9, 329)	59 (11, 329)	74 (10, 154)	86 (15, 314)	63 (9, 296)
Subjects evaluable for target-tumor response****; [# with genetic profile available]	71 [64]	24 [21]	22 [21]	7 [7]	9 [8]

* In addition to the subjects shown, 4 subjects with HNSCC were enrolled; no SCCA subjects were enrolled
** Includes subjects in the Dose Escalation phase concurrently enrolled in Cohort Expansion (3 CRC; 1 HGSOC; 1 mCRPC)
*** Prior radiation therapy regimens: n=44
**** Subjects with pre- and post-treatment target tumor measurements who received ≥ 75% of total planned C1 dose at ≥ 300mg, or continued on-study after 2 cycles of treatment at any dose level
Data cut off: 03 May 2019

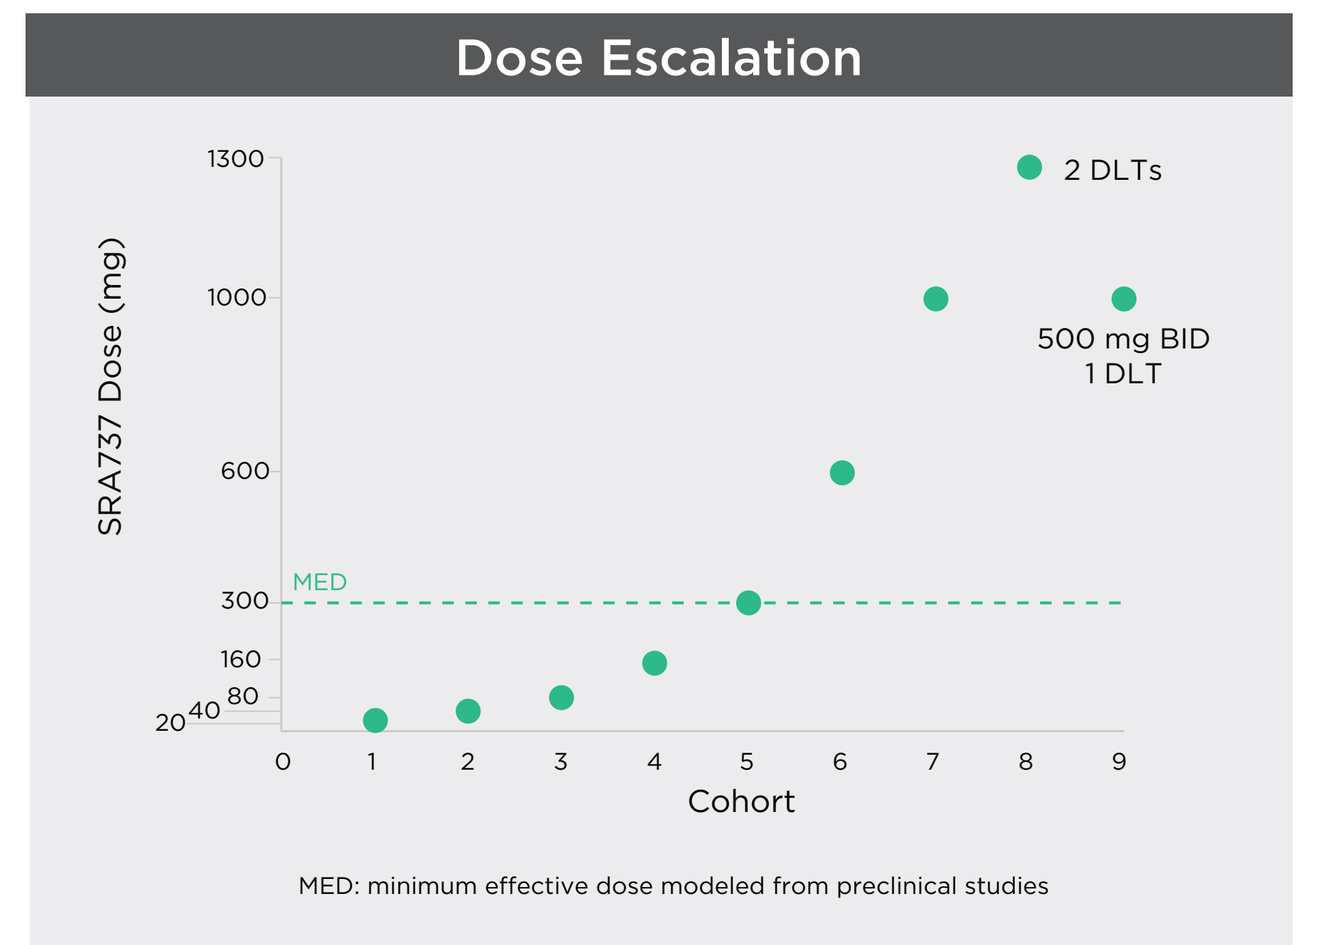

Dose Escalation
SRA737 Dose (mg)
2 DLTs
500 mg BID
1 DLT
MED
Cohort
MED: minimum effective dose modeled from preclinical studies

The plasma pharmacokinetics of SRA737 were reproducible and generally dose concordant. The C_{min} (289 ng/mL) at 1000 mg exceeded that determined in preclinical efficacy models to have anti-tumor activity (100 nM; ca 37.9 ng/mL). All doses ≥ 300 mg QD also exceeded this threshold level.

	Dose (mg)	Tmax* (h)	Cmax (ng/mL)	Cmin (ng/mL)	AUC0-24h (ng•h/mL)	t1/2 (h)	CL/F (L/h)	Vz/F (L)
Day -7 to -4	160 (n=1)	4 (-)	178 (-)	31.7 (-)	1,618 (-)	13.8 (-)	70.0 (-)	1,392 (-)
	300 (n=1)	4 (-)	353 (-)	41.2 (-)	3,152 (-)	8.6 (-)	81.9 (-)	1,015 (-)
	600 (n=4)	2 (1 - 2)	998 (337)	129 (49.4)	8,870 (3,170)	12.9 (1.4)	56.9 (20.8)	1,090 (516)
	800 (n=38)	4 (1 - 6)	1,884 (606)	234 (98.2)	18,537 (6,056)	10.5 (1.8)	39.1 (14.9)	596 (281)
	1000 (n=44-45)	4 (0.5 - 8)	2,098 (805)	289 (151)	20,270 (7,656)	11.0 (1.7)	44.9 (22.5)	704 (398)
	1300 (n=3)	2 (2 - 4)	3,228 (1,320)	466 (234)	29,796 (11,233)	11.8 (3.1)	38.6 (19.7)	625 (221)

* Tmax reported as Median (min-max)

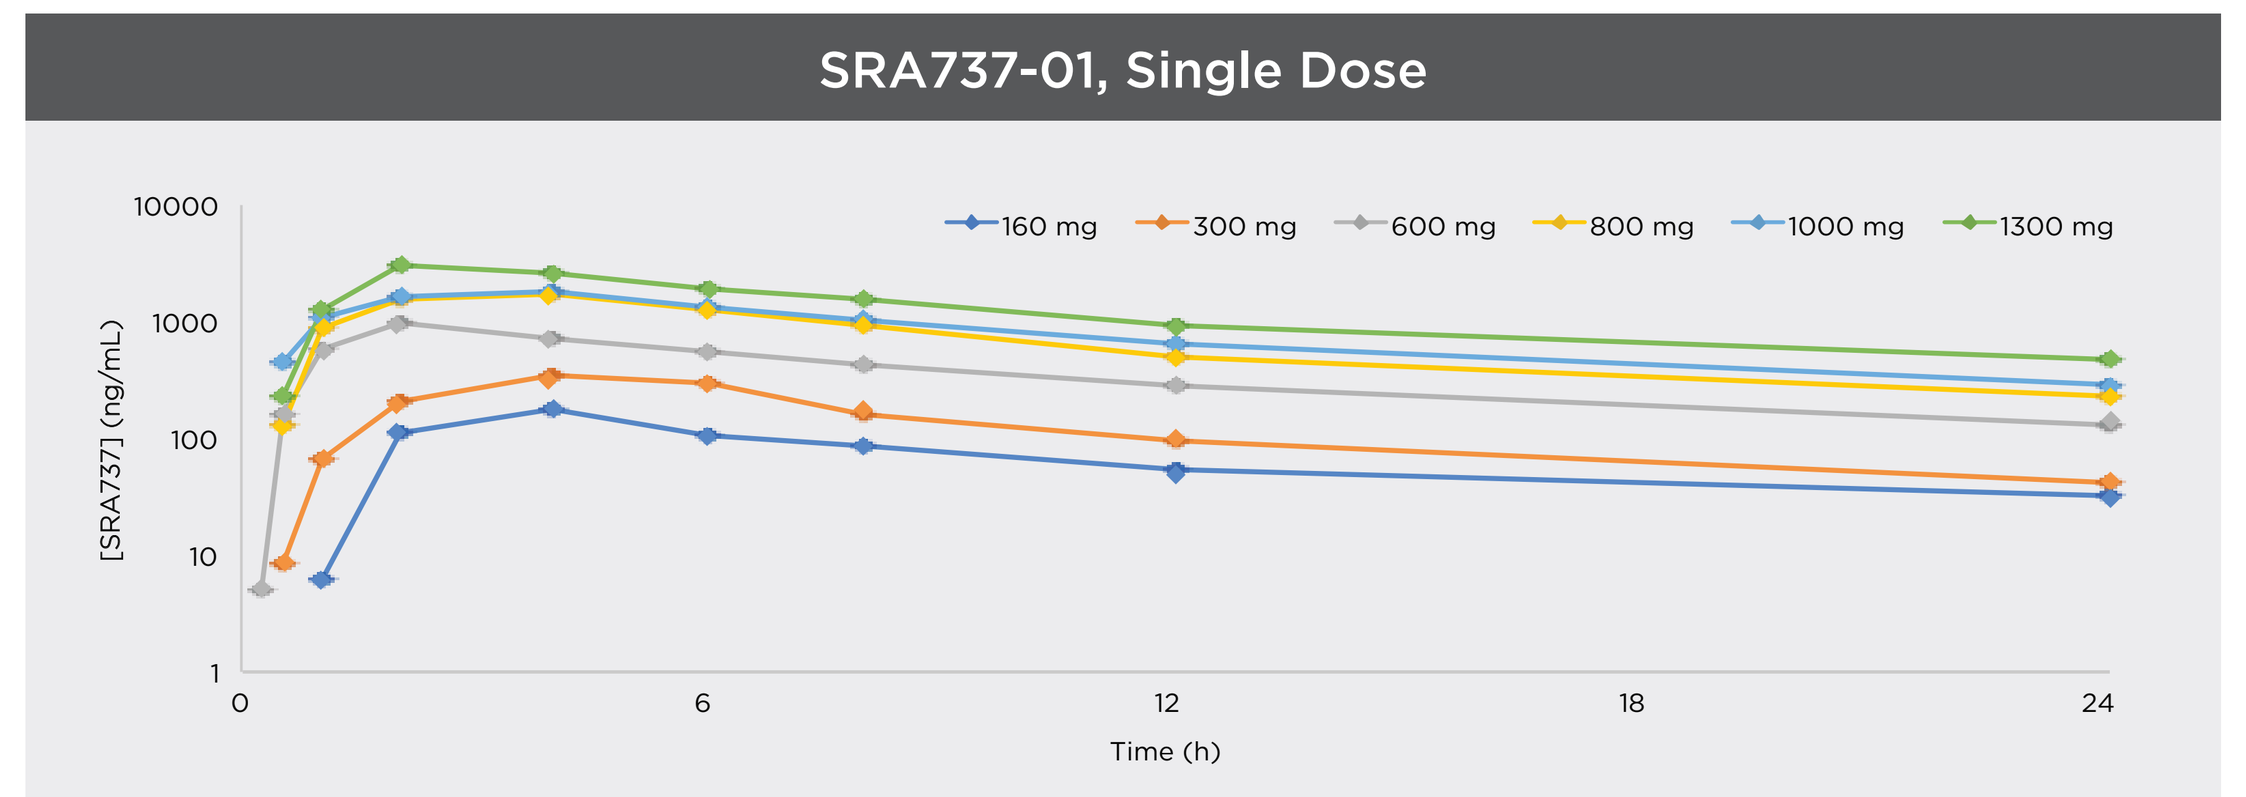

SRA737-01, Single Dose
160 mg
300 mg
600 mg
800 mg
1000 mg
1300 mg
[SRA737] (ng/mL)
Time (h)

Safety

- Treatment-emergent adverse events (TEAEs) were reported in 106 (99%) subjects and 97 subjects (91%) experienced at least one SRA737-related event.
- The majority of TEAEs were mild to moderate in severity (90% Grade 1/Grade 2).
- The most common TEAEs were diarrhea (68%), nausea (66%), vomiting (51%) and fatigue (47%).
- The most common ≥ Grade 3 TEAEs were neutropenia and disease progression (8% each), lymphocyte count decreased (5%), infection and hyponatremia (4% each) and nausea, abdominal pain, abdominal pain upper, fatigue, AST increased, dyspnea, pleural effusion, and rash maculo-papular (3% each).
- The most common ≥ Grade 3 SRA737-related TEAEs were neutropenia (8%) and rash maculo-papular (3%).
- Six Grade 5 TEAEs were reported up to 30-days post last treatment; none were considered related to SRA737.
- Median duration of exposure was 2.3 cycles (range < 1 to 9 cycles).
- No evidence of emergent or cumulative toxicity and/or declining tolerability was observed with up to 9 cycles of SRA737 administered.

Treatment-Emergent Adverse Events (TEAEs)	Occurring in ≥ 20% of subjects N=107, n (%)	≥ Grade 3 n (%) N=107, n (%)
Subjects with any TEAE	106 (99.1%)	73 (68.2%)
Diarrhea	73 (68.2%)	2 (1.9%)
Nausea	71 (66.4%)	3 (2.8%)
Vomiting	55 (51.4%)	1 (0.9%)
Fatigue	50 (46.7%)	3 (2.8%)
Decreased appetite	25 (23.4%)	0
Anemia	23 (21.5%)	2 (1.9%)

TEAEs regardless of the investigator's assessment of causality;
Data cut off: 23 March 2019.

Results

- 107 subjects received treatment with SRA737 across both escalation and expansion cohorts. In the Cohort Expansion phase, the largest number of subjects were enrolled in the HGSOC cohort (n=38) with the next largest number in the CRC cohort (n=27).
- **Evidence of anti-tumor activity** was observed in subjects with HGSOC, colorectal, prostate and non-small cell lung cancer; no RECIST PRs or CRs were confirmed, but several noteworthy tumor reductions were recorded.
- HGSOC appeared as the most sensitive tumor to SRA737 monotherapy in the SRA737-01 study **(Figure 3)**. Although heavily pre-treated (~5 prior lines), the HGSOC cohort demonstrated **directionally favorable disease control** (DCR = 54%) with notable maximal tumor reductions of 29% and 27% observed in two patients.
- At the time of the data cut off, **durable stable disease (SD) lasting ≥ 4 months** was recorded in 22 (21%) subjects and was observed in all Cohort Expansion tumor types (except HNSCC; 4 subjects) **(Figure 2)**.
- In keeping with the signal seeking objective of this study, tumor responses were further examined with respect to the genetic profiles determined for the tumor types enrolled and treated **(Figure 4)**.
- Among gene networks with > 10 subjects, alterations in several genes correlated either positively, or negatively (RAS; **Figure 5**), with response to SRA737 **(Table 1)** across multiple indications.
- Notably, genetic alterations in multiple components of the **FA/BRCA gene network** were associated with a 71% DCR and DOS of 3.8 cycles **(Figure 7)**. The majority of subjects with notable responses (tumor reduction and/or > 4 months SD) harbored alterations in either or both the **PI3K (Figure 6)** and **FA/BRCA** gene networks **(Table 2)**.
- Results data cut off: 03 May 2019; Data not final

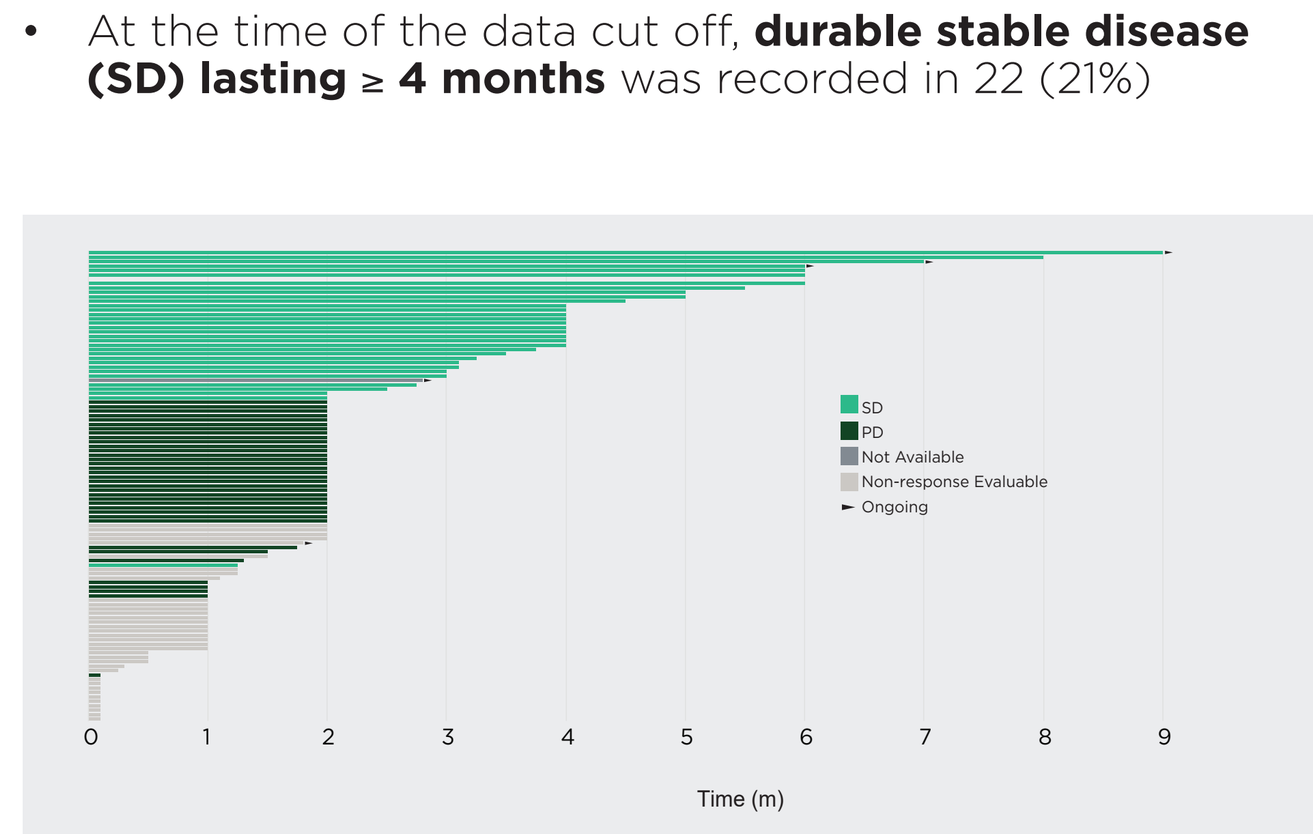

SD
PD
Not Available
Non-response Evaluable
Ongoing
Time (m)

Figure 2. Duration on Treatment in SRA737-01. Data shown represent the duration on treatment for all subjects (N=107). A best overall response of SD was seen in 34 (32%) subjects; several subjects (21%) achieved durable SD ≥ 4 months. The median treatment delay from consent to C1D1 to allow for genetic profiling exceeded 2 months, highlighting the challenge of prospective genetic screening using tumor tissue. In a population of subjects with advanced disease (4+ prior lines), this delay in treatment initiation arguably exacerbates underlying disease progression, and is reflected in early drop-outs on study. The identification of both positive- and negative-selection genetic markers as determined in this clinical study may provide a focused ctDNA enrichment strategy for deployment in future clinical studies, potentially expediting prospective enrollment.

Functional Gene Category	Gene Network	Gene	Number of Subjects	Number of RAS Wild-Type Subjects	DCR‡ (%)
Cell Cycle Dysregulation	p53 Pathway	*MDM2*, *TP53*	50	33	56%
	G1/S	*RB1*, *CDKN1A/B*, *CDKN2A/B/C*	7	5	40%
Oncogenic Drivers	CCNE	*CCNE1*, *FBXW7*, *PARK2*	11	9	67%
	RAS	*KRAS*, *NRAS*, *HRAS*	20	NA	25%
	MYC	*MYC*, *MYCN*, *MYCL1*	8	4	50%
	PI3K	*PIK3CA*, *PTEN*, *AKT1/2/3*	19	13	77%
DNA Damage Response and Repair Network	FA/BRCA	*CDK12*, *FANC**, *RAD***, *ATR*, *BRCA1/2*, *RAD51B*, *RAD51C*, *PRKDC*	22	21	71%
	HR/NHEJ	*BRCA1/2*, *RAD51B*, *RAD51C*, *PRKDC*, *PALB2*, *ATM*	19	19	68%
	Chromatin	*MLL2*, *ARID1A*, *ARID1B*	6	3	100%
	Mismatch Repair	*MLH1*, *MSH2*, *MSH6*, *PMS2*	4	4	75%
	DNA Pol	*POLD1*, *POLE*	1	1	0%

‡ Rate for RAS Wild-Type subjects
*Includes FANC A, C, and G
**Includes RAD51 and RAD51C

Table 1. Disease Control Rates Vary Across Defined Gene Networks.

RS-driver genes encompassing functional categories (G1/S, Oncogenes, DNA repair genes) were surveyed to identify gene networks and/or individual genes that enriched for sensitivity to treatment with SRA737. Mutations in the RAS gene network were associated with relatively poor response. Alterations in PI3K and FA/BRCA gene networks were associated with favorable disease control. A directionally positive correlation with CCNE is suggested, albeit CCNE network alterations were observed in only a limited number of subjects.

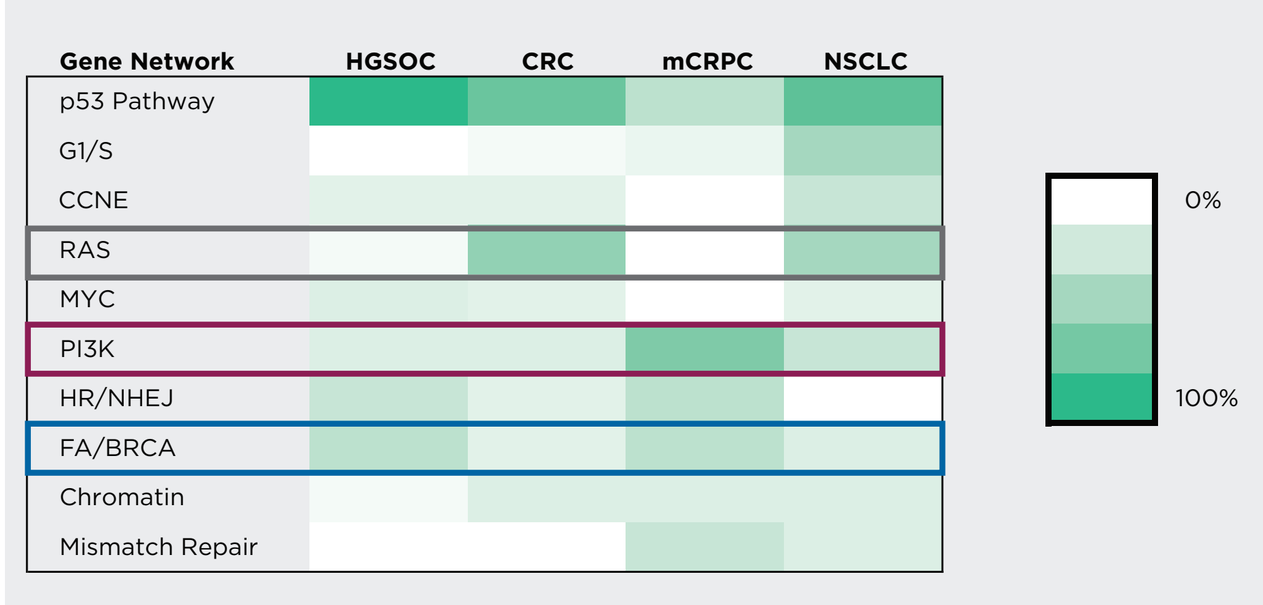

Gene Network
HGSOC
CRC
mCRPC
NSCLC
p53 Pathway
G1/S
CCNE
RAS
MYC
PI3K
HR/NHEJ
FA/BRCA
Chromatin
Mismatch Repair
0%
100%

Figure 4. Frequency of Gene Network Alterations Across Indications. The heatmap displays the frequency of observed gene network alterations observed in SRA737-01 across treatment cohorts, represented by percent of subjects with gene alterations.

Tumor Type	Best % Change	Duration on Treatment (Cycles)	# Lines Prior Therapy	Gene Network: FA/BRCA	CCNE	PI3K
HGSOC	-29%	4	5			
HGSOC	-27%	2	4			
mCRPC	-21%	4	5			
CRC	-19%	8	3			
HGSOC	-19%	6	6			
CRC	-14%	6	2			
HGSOC	-12%	6	3			
NSCLC	-6%	6▶	3			
mCRPC	0%	9▶	2			
mCRPC	2%	7▶	5			
CRC	4%	6	2			
NSCLC	4%	5	3			
CRC	8%	6	3			

▶ Ongoing

Table 2. Summary of Subjects with Notable Responses and their Respective Gene Network Alterations.

Subjects who achieved SD of ≥ 4 cycles and/or best % tumor decrease of > 10% are correlated with their respective gene network alterations. CCNE gene network alterations (DCR=67%) partially overlapped with FA/BRCA subjects but was mutually exclusive with PI3K network alterations.

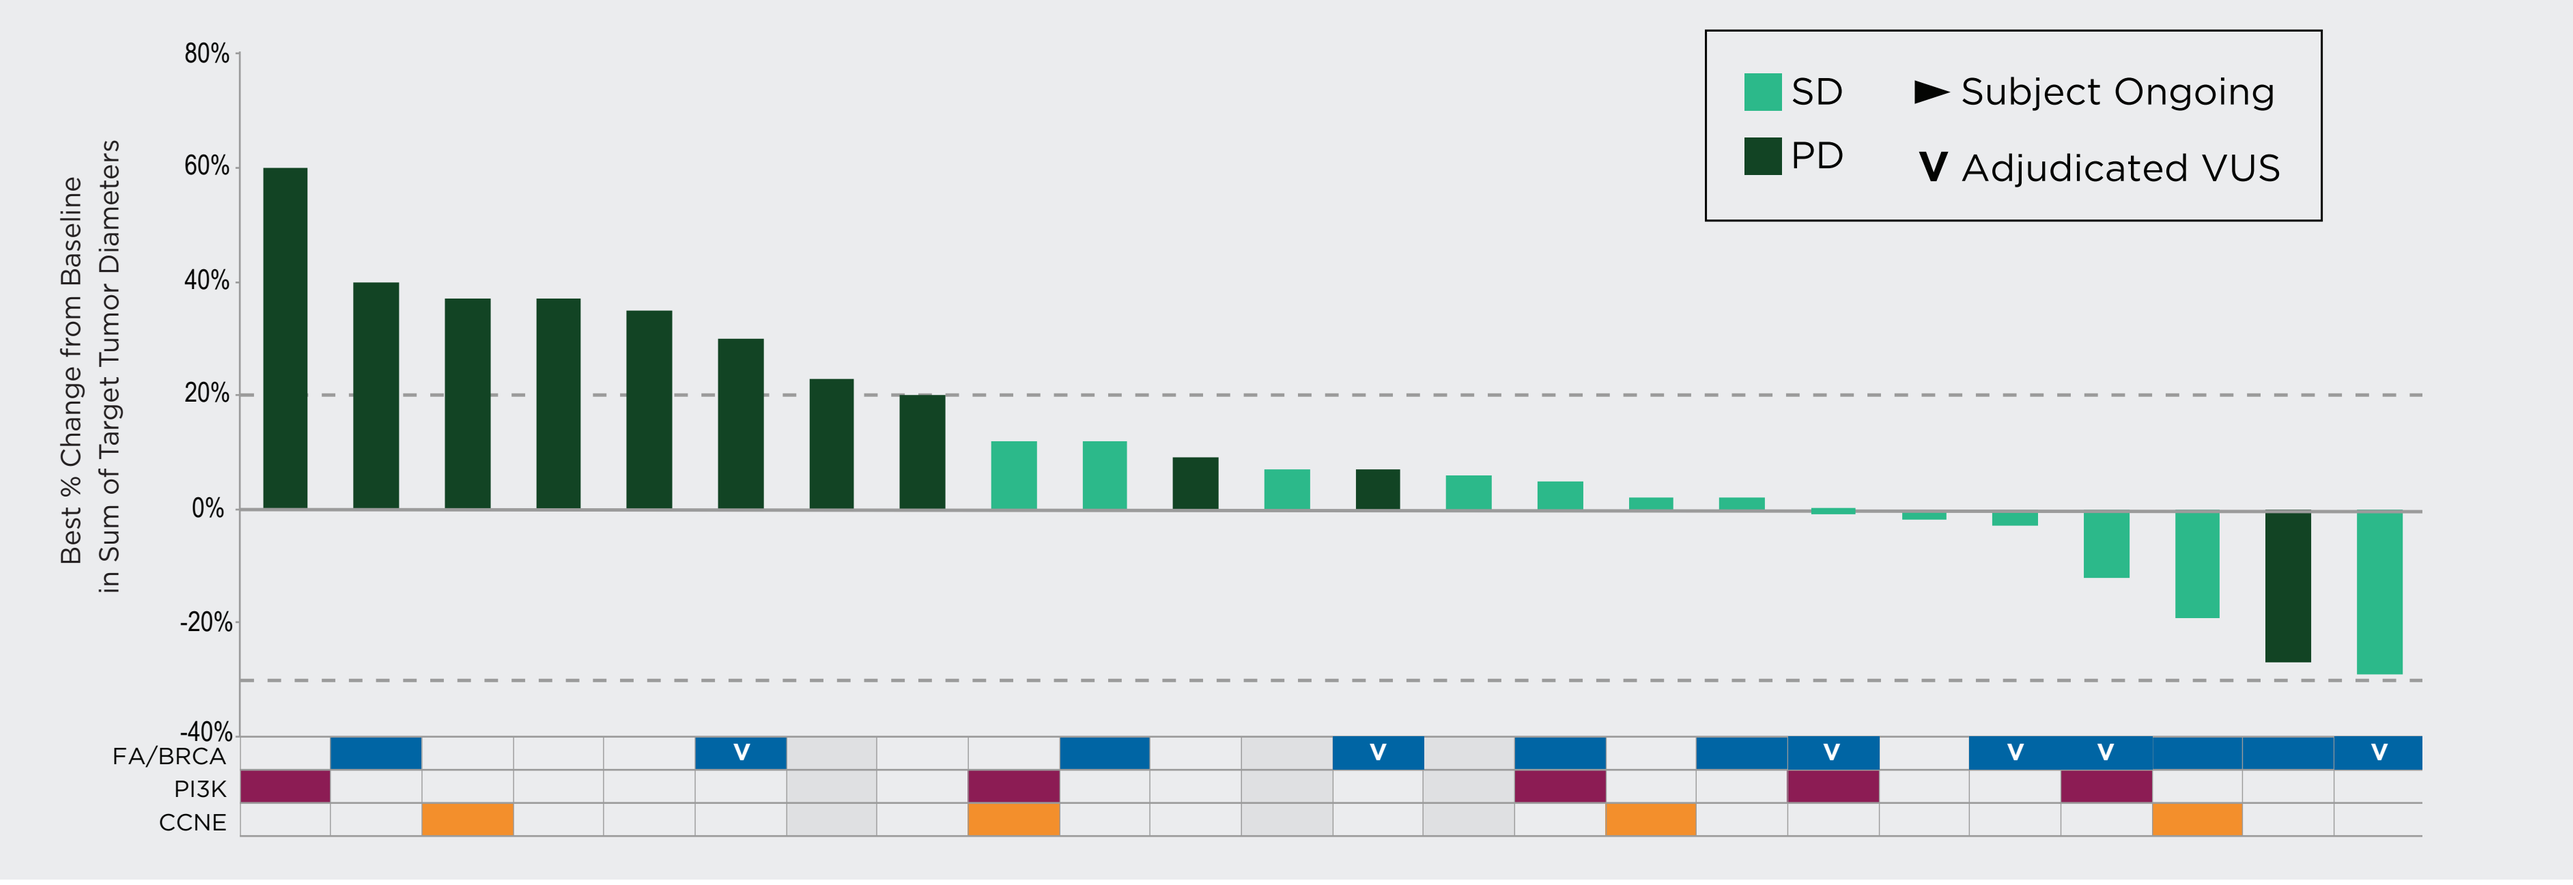

SD
PD
Subject Ongoing
V Adjudicated VUS
Best % Change from Baseline in Sum of Target Tumor Diameters
FA/BRCA
PI3K
CCNE

Figure 3. Directionally Positive HGSOC Cohort Responses.

HGSOC represented the most sensitive indication identified in this study. FA/BRCA gene network alterations were enriched in the HGSOC cohort, particularly in subjects with tumor reduction. Although a cohort of subjects was specifically enrolled to putatively enrich for CCNE network alterations, genetic analysis revealed that only 4/24 subjects had tumors harboring unambiguously positive *CCNE1* amplification, resulting in 3 SD and 1 progressive disease (PD). Albeit a very limited dataset, no clear correlation with sensitivity was observed in association with *CCNE1* amplification.

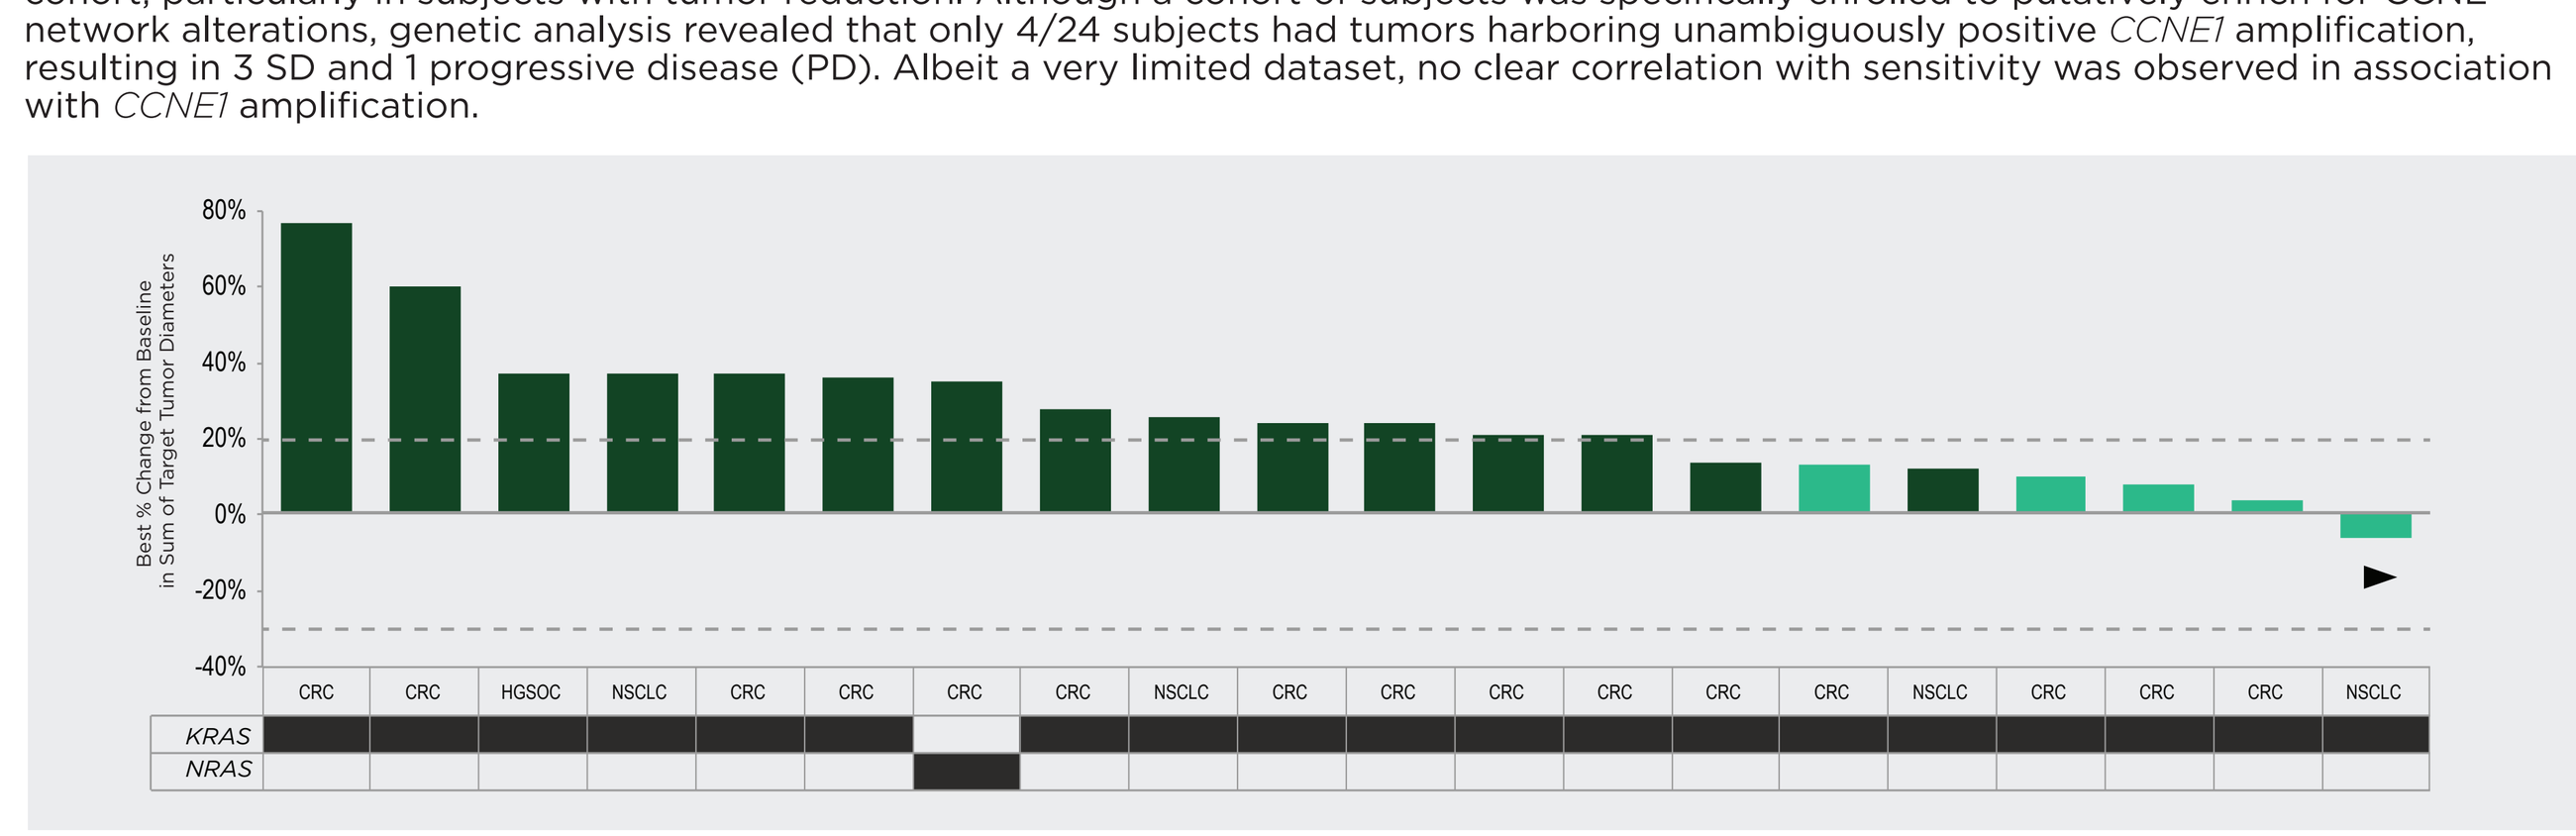

Best % Change from Baseline in Sum of Target Tumor Diameters
KRAS
NRAS

Figure 5. RAS Gene Network Alterations May be Antagonistic to SRA737 Sensitivity.

Mutations in *KRAS* and *NRAS* trended toward poor DCR (25%) with 15/20 PDs and an overall average tumor % change of +20%, as well as short duration on study (DOS) of 2 cycles. Given the strong negative correlation with activity, additional pathway/signal searching analyses was performed with exclusion of RAS mutated tumors.

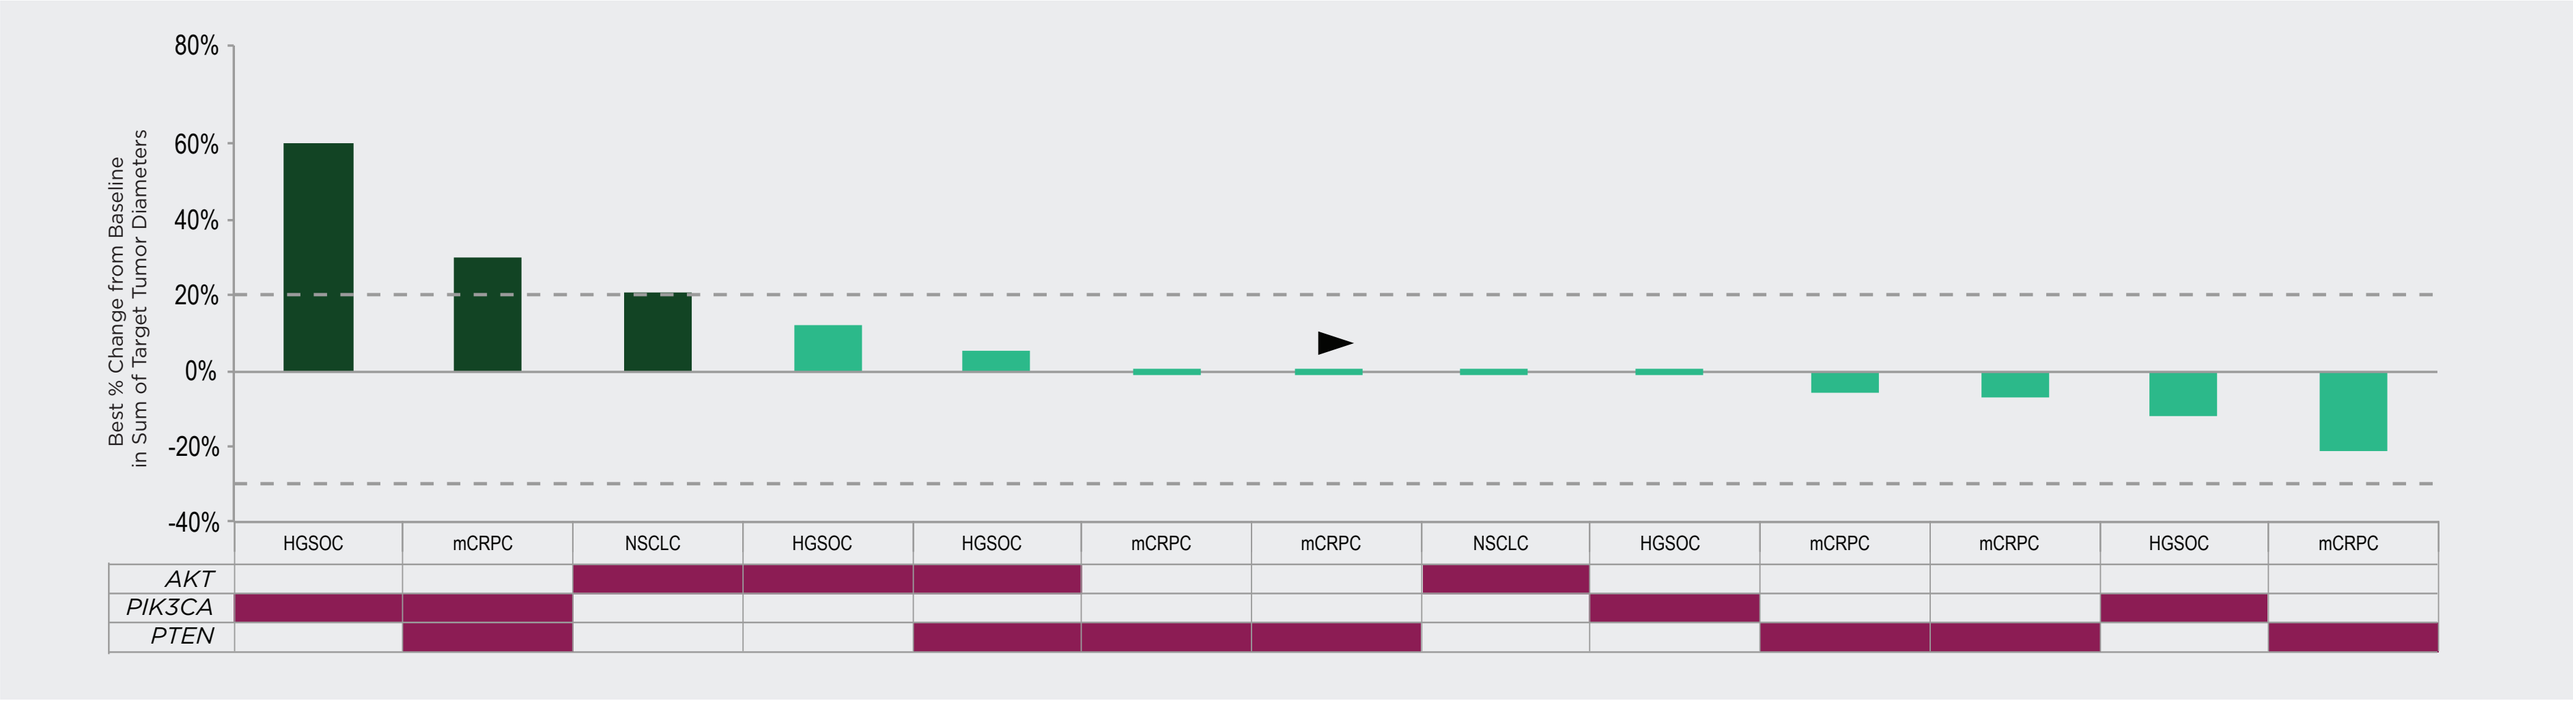

Best % Change from Baseline in Sum of Target Tumor Diameters
AKT
PIK3CA
PTEN

Figure 6. PI3K Gene Network Alterations May Enhance SRA737 Sensitivity.

Alterations in the PI3K gene network (*PIK3CA, AKT, PTEN*) were associated with a 77% DCR.

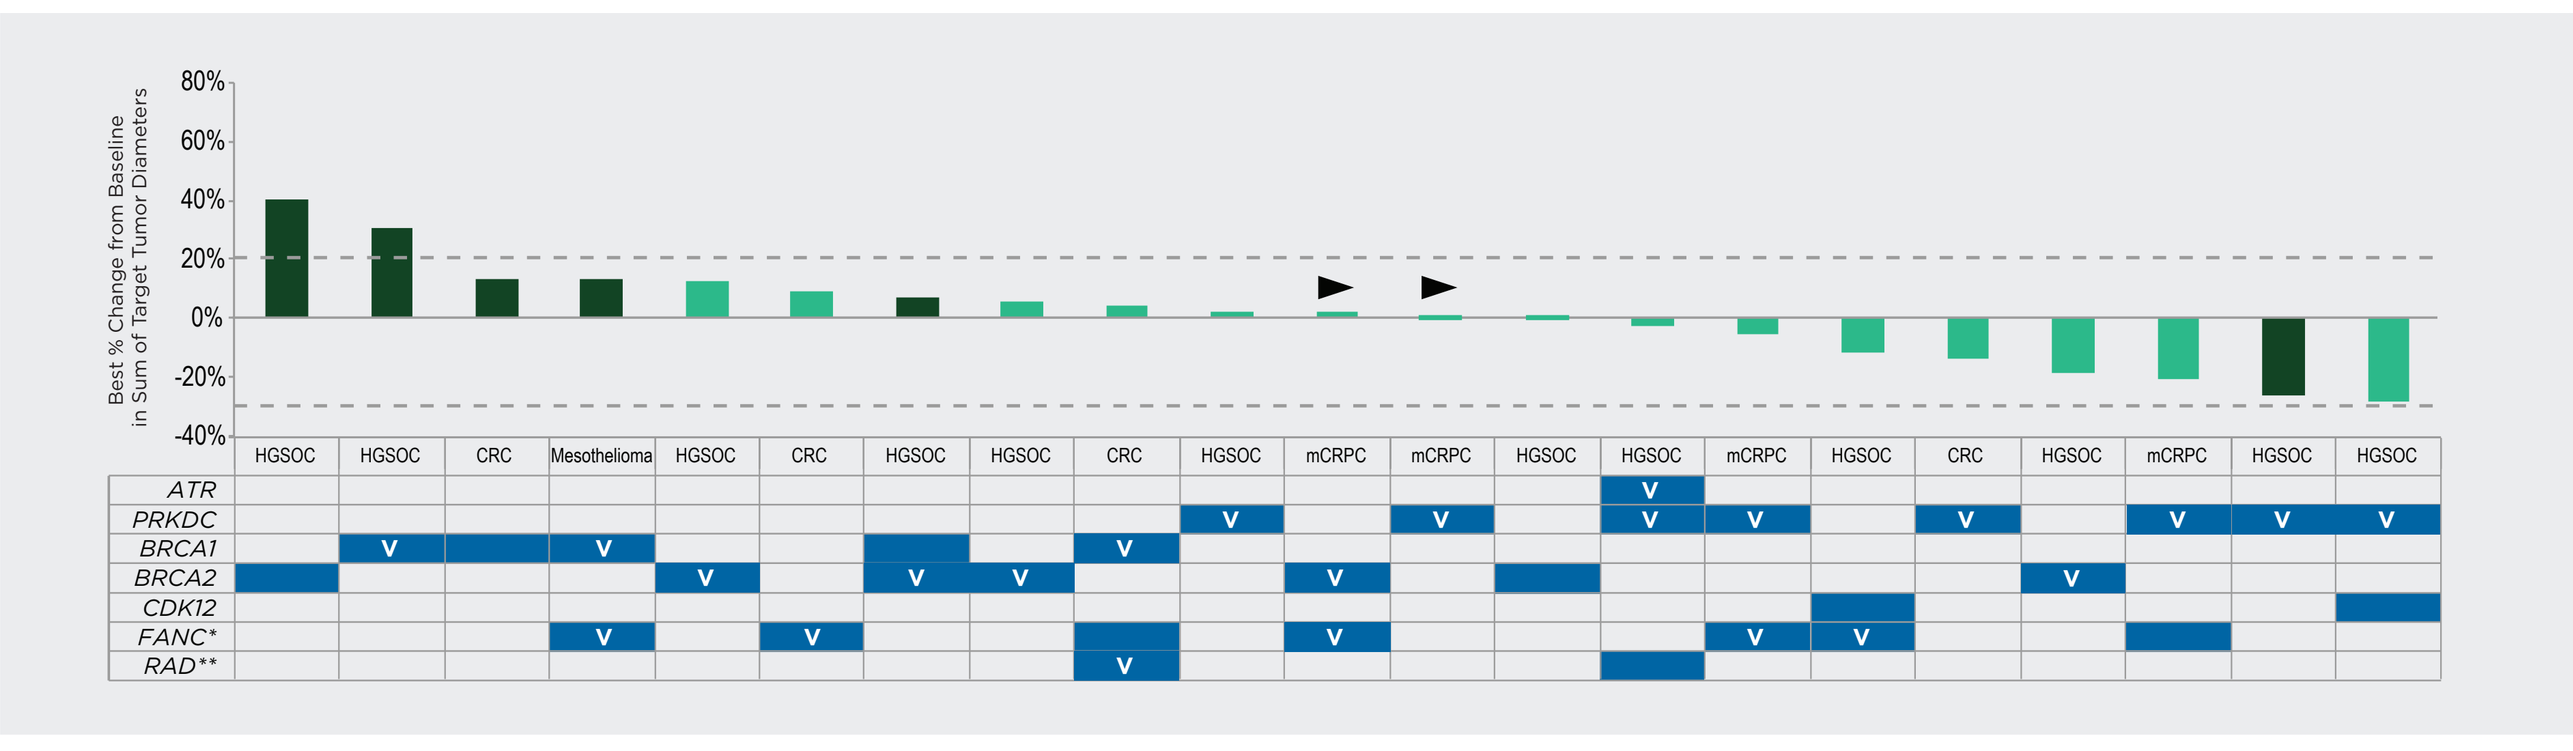

Best % Change from Baseline in Sum of Target Tumor Diameters
ATR
PRKDC
BRCA1
BRCA2
CDK12
FANC*
RAD**

Figure 7. FA/BRCA Replication Fork Gene Network Alterations May Enhance SRA737 Activity.

Many of the subjects demonstrating tumor decrease in SRA737-01 harbored one or more genetic alterations in the FA/BRCA gene network, frequently including a secondary mutation in a DDR kinase gene (*ATR, PRKDC*).

Conclusions

- Overall, these data provide **promising evidence of SRA737 anti-tumor activity** and identify several gene networks associated with enhanced SRA737 sensitivity.
- In this first-in-human trial of SRA737 monotherapy, the MTD was 1000 mg/day, and based on overall tolerability and PK, the recommended monotherapy dose is 800 mg/day. **These results highlight the safety and tolerability of SRA737**.
- The signal-seeking study surveyed broadly across tumor indications and tumor RS-driver genetics to identify potential SRA737-sensitive contexts. Preliminary **evidence of anti-tumor activity** observed in subjects with HGSOC, colorectal, prostate and non-small cell lung cancer suggests several intrinsic sources of RS contribute to or enhance Chk1 sensitivity.
- The heavily pre-treated HGSOC cohort (~5 prior lines) demonstrated **directionally favorable disease control (DCR=54%)** with notable maximal tumor reductions of 29% and 27% in two subjects. No clear trend toward enhanced sensitivity was noted with *CCNE1* gene amplification; the small subset of subjects enrolled with unambiguous *CCNE1* amplifications renders definitive conclusions challenging.
- An evaluation of i) notable tumor volume reductions and ii) longest DOS, revealed that subjects whose tumors harbored **FA/BRCA network mutations** displayed the most favorable outcomes (DCR=71%; DOS=3.8 cycles). The FA/BRCA gene network encodes a series of Fanconi Anemia and other proteins involved directly or indirectly in replication fork metabolism and management of RS.
- Importantly, these sensitivity trends were observed across multiple indications, suggesting a potential histology-independent sensitization. Similar observations were made in a clinical study of SRA737+low dose gemcitabine (LDG) (NCT02797977).
- Within the FA/BRCA pathway, several notable tumor volume decreases occurred in subjects harboring two genetic alterations, such as a DDR checkpoint kinase gene or PI3K network. This genetic sensitivity correlation, which was also observed in the SRA737+LDG clinical study, suggests that multiple mutations may lead to elevated intrinsic RS and genomic instability, and/or be a consequence thereof.
- These findings suggest that additional RS, such as **via extrinsic sources**, may be necessary to generate durable objective responses with a highly selective Chk1i, as evidenced by anti-tumor activity demonstrated in the SRA737-02 clinical study, where potentiating LDG was combined with SRA737.

Information
For more information, email info@sierraoncology.com or visit www.sierraoncology.com

Acknowledgments
This study was sponsored by Sierra Oncology. We would like to thank all participating patients and their families. Investigators thank Cancer Research UK, the Experimental Cancer Medicine Centre (ECMC) and the National Institute of Health Research for research infrastructure support.